Poster ID: 40

Efficacy of the Oral Plasma Kallikrein Inhibitor Sebetralstat (KVD900) by Attack Location in a Phase 2 Clinical Trial in Patients With Hereditary Angioedema

Emel Aygören-Pürsün,[1] Andrea Zanichelli,[2] Danny M. Cohn,[3] Paul K. Audhya,[4] Peter Williams,[5] Chris M. Yea,[4] Michael D. Smith[4]

[1]University Hospital Frankfurt, Frankfurt, Germany; [2]ASST Fatebenefratelli Sacco, Ospedale Luigi Sacco-University of Milan, Milan, Italy; [3]Amsterdam University Medical Centers, University of Amsterdam, Amsterdam, Netherlands; [4]KalVista Pharmaceuticals, Salisbury, UK, and Cambridge, MA, US; [5]Veramed Limited, Twickenham, UK

Background

- Hereditary angioedema (HAE) is a rare and potentially life-threatening genetic disease involving abnormal functioning of the kallikrein-kinin system leading to increased vascular permeability and characterized by unpredictable, recurrent, and often painful episodes of swelling of varying severity and location[1-4]
 - Peripheral (also called subcutaneous) attacks affecting the hands, feet, and genitourinary tract are the most common, and may result in disability for 1-5 days[2,3]
 - Abdominal (also called submucosal) attacks are characterized by discomfort, distension, and nausea that can sometimes progress to severe pain with vomiting and diarrhea[2,3]
- Treatment guidelines for HAE recommend that patients have access to medications for on-demand treatment of HAE attacks and treat every attack as early as possible[5-7]
 - Currently, all approved on-demand treatments require parenteral administration, which presents significant challenges with time needed for medication preparation, administration, and injection-site−associated pain and discomfort[8-11]
 - There remains an unmet need for a safe and effective oral on-demand treatment option for HAE attacks regardless of attack location or severity
- Sebetralstat is a novel investigational oral plasma kallikrein inhibitor for the on-demand treatment of HAE attacks[12]
- In a phase 2 randomized clinical trial, a single oral dose of sebetralstat 600 mg was effective in slowing the progression of HAE attacks and was generally safe and well tolerated[12]
- This post hoc analysis of data from the phase 2 trial assessed achievement of and time to symptom relief, improvement, attack resolution and use of rescue medication by attack location

Methods

- Trial design (NCT04208412) is shown in **Figure 1**
- Enrolled patients were aged ≥18 years with HAE type I or II who had ≥3 attacks in the past 93 days and were not on prophylactic therapy
- In the crossover part of the trial (part 2), patients were randomized to treat 2 mild to moderate HAE attacks (severe attacks and orofacial-pharyngeal-laryngeal attacks were excluded) with sebetralstat 600 mg or placebo in 1 of 2 sequences
- Attacks that involved the face or larynx were not eligible for treatment with the study drug

Figure 1. Trial Design

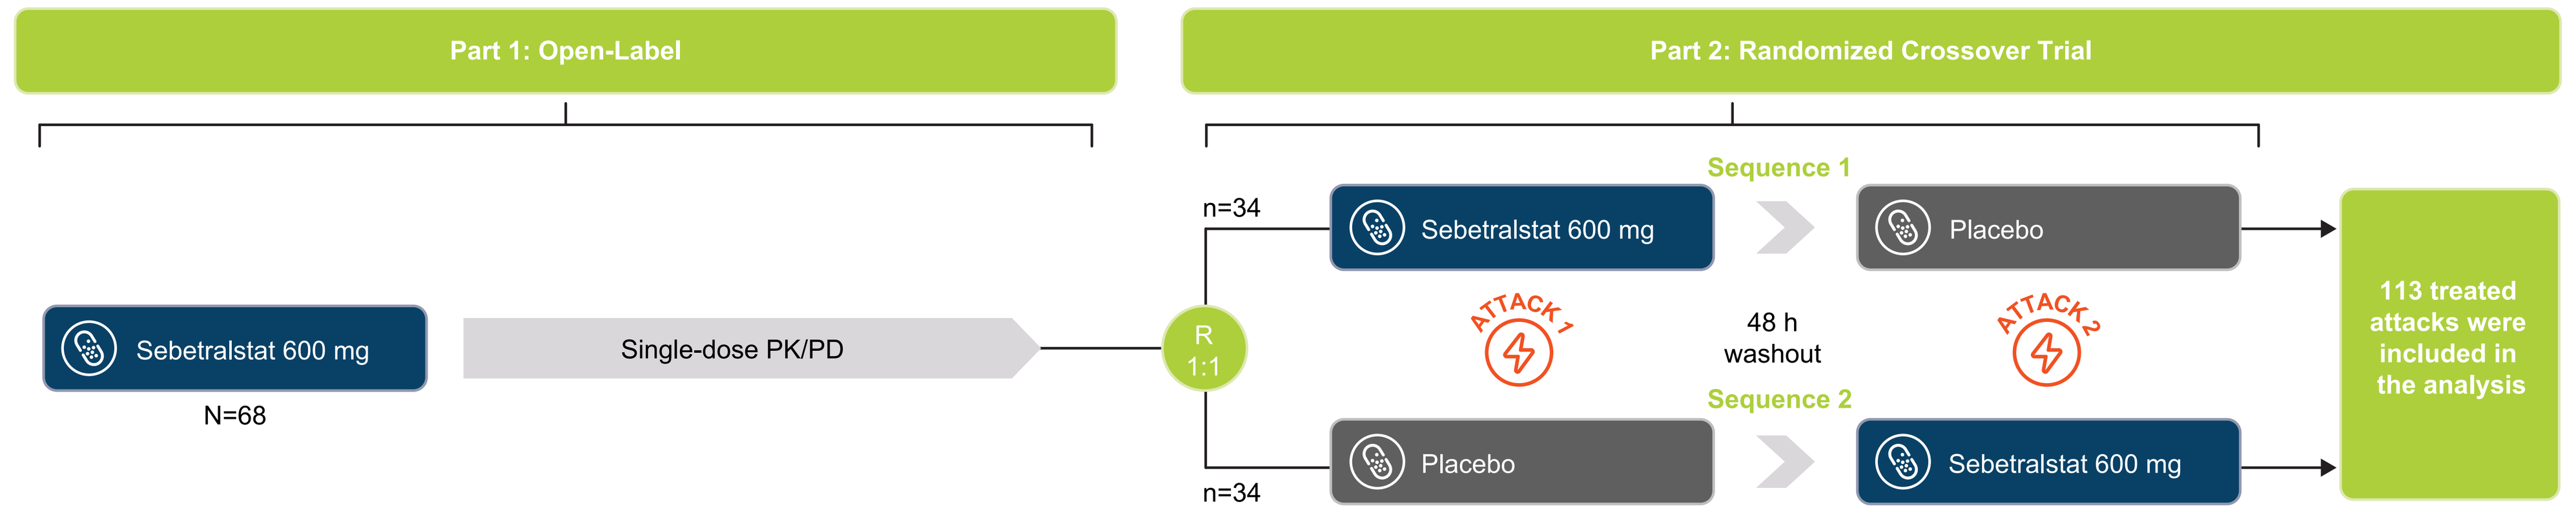

Part 1: Open-Label
Part 2: Randomized Crossover Trial
Sebetralstat 600 mg
N=68
Single-dose PK/PD
R
1:1
n=34
n=34
Sequence 1
Sebetralstat 600 mg
Placebo
ATTACK 1
48 h
washout
ATTACK 2
Sequence 2
Placebo
Sebetralstat 600 mg
113 treated attacks were included in the analysis

h, hour; PD, pharmacodynamic; PK, pharmacokinetic; R, randomized.

Classification of Attacks

- For this analysis, attacks were categorized as
 - abdominal: included abdominal symptoms at attack onset (with or without peripheral symptoms)
 - peripheral: included only peripheral (and no abdominal) symptoms at attack onset

Outcome Measures

- Symptom relief was defined as a score of "A Little Better" or higher for 2 consecutive timepoints within 12 hours of study drug administration on the Patient Global Impression of Change (PGI-C) scale
- Baseline attack severity was evaluated by Patient Global Impression of Severity (PGI-S) scale (numeric values from 0 to 4 represented categorical PGI-S scores from "None" to "Very Severe") and by individual component (abdominal pain, skin pain, skin swelling) and composite (mean of the components) scores on a 100-mm visual analog scale (VAS)
- Improvement on PGI-S scale was defined as improvement from baseline by 1 or more levels within 12 hours
- Attack resolution was defined as PGI-S score of 0 ("None") within 24 hours:
 - Attack resolution was also defined as a score <10 mm for all VAS components for 3 consecutive timepoints within 24 hours; attacks where all 3 VAS components were <10 mm at baseline were excluded from the analysis
- Use of rescue medication within 12 and 24 hours was assessed by attack location
- Results are presented using descriptive statistics; symptom evaluation scales are shown in **Figure 2**

Figure 2. Symptom Evaluation Scales

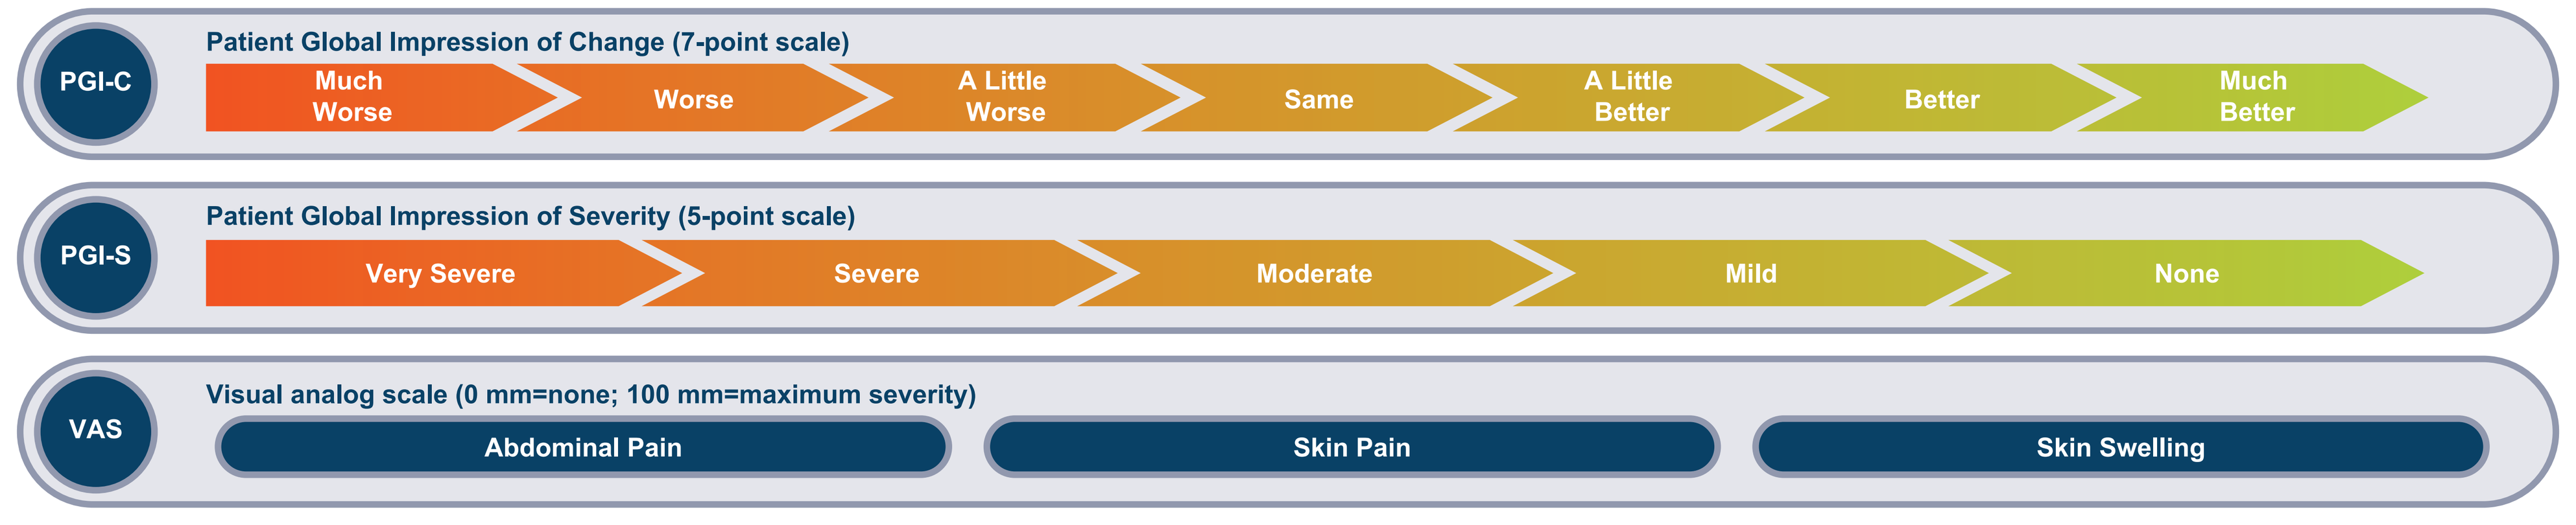

PGI-C
Patient Global Impression of Change (7-point scale)
Much Worse
Worse
A Little Worse
Same
A Little Better
Better
Much Better
PGI-S
Patient Global Impression of Severity (5-point scale)
Very Severe
Severe
Moderate
Mild
None
VAS
Visual analog scale (0 mm=none; 100 mm=maximum severity)
Abdominal Pain
Skin Pain
Skin Swelling

PGI-C, Patient Global Impression of Change; PGI-S, Patient Global Impression of Severity; VAS, visual analog scale.

Results

Attack Characteristics

- 113 attacks were treated in the trial, of which 58 were treated with sebetralstat
- Of sebetralstat-treated attacks, 18 (31.0%) were categorized as abdominal and 40 (69.0%) as peripheral
 - Of the attacks classified as abdominal, 2 (11.1%) attacks included both abdominal and peripheral symptoms (abdomen and genitals; abdomen and arm)
 - Peripheral attacks most commonly occurred in the arms or legs (62.5%) followed by the genitourinary tract (15.0%), hand (12.5%), and other locations (eg, buttock, shoulder, foot) (10.0%)

Baseline Attack Severity

- There was a tendency for abdominal attacks to be rated as more severe than peripheral attacks at baseline (**Figure 3**)

Figure 3. Baseline Severity of Attacks (PGI-S and Composite VAS) by Attack Location

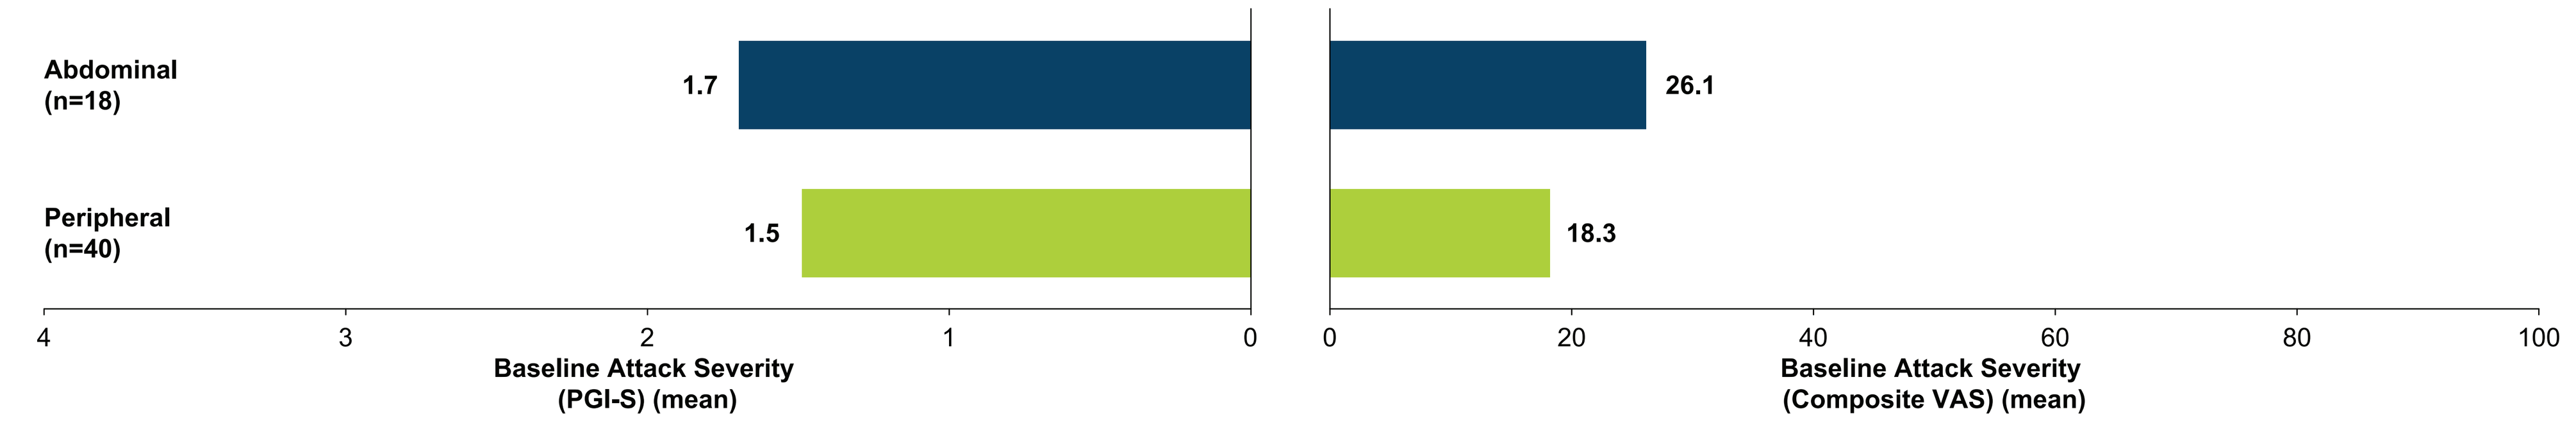

Abdominal (n=18)
Peripheral (n=40)
1.7
1.5
26.1
18.3
4
3
2
1
0
Baseline Attack Severity (PGI-S) (mean)
0
20
40
60
80
100
Baseline Attack Severity (Composite VAS) (mean)

PGI-S, Patient Global Impression of Severity; VAS, visual analog scale.

- Mean VAS component scores at baseline by attack location
 - Skin pain: 13.8 (abdominal), 18.0 (peripheral)
 - Skin swelling: 23.2 (abdominal), 34.9 (peripheral)
 - Abdominal pain: 41.2 (abdominal), 2.2 (peripheral)

Symptom Relief

- 83.3% of attacks in abdominal and 80.0% in peripheral locations achieved symptom relief as assessed on PGI-C within 12 hours (**Figure 4, left panel**)
- Median time to symptom relief (PGI-C) within 12 hours was slightly shorter for abdominal attacks compared with peripheral attacks (**Figure 4, right panel**)

Figure 4. Achievement of Symptom Relief and Time to Symptom Relief (PGI-C) Within 12 Hours by Attack Location

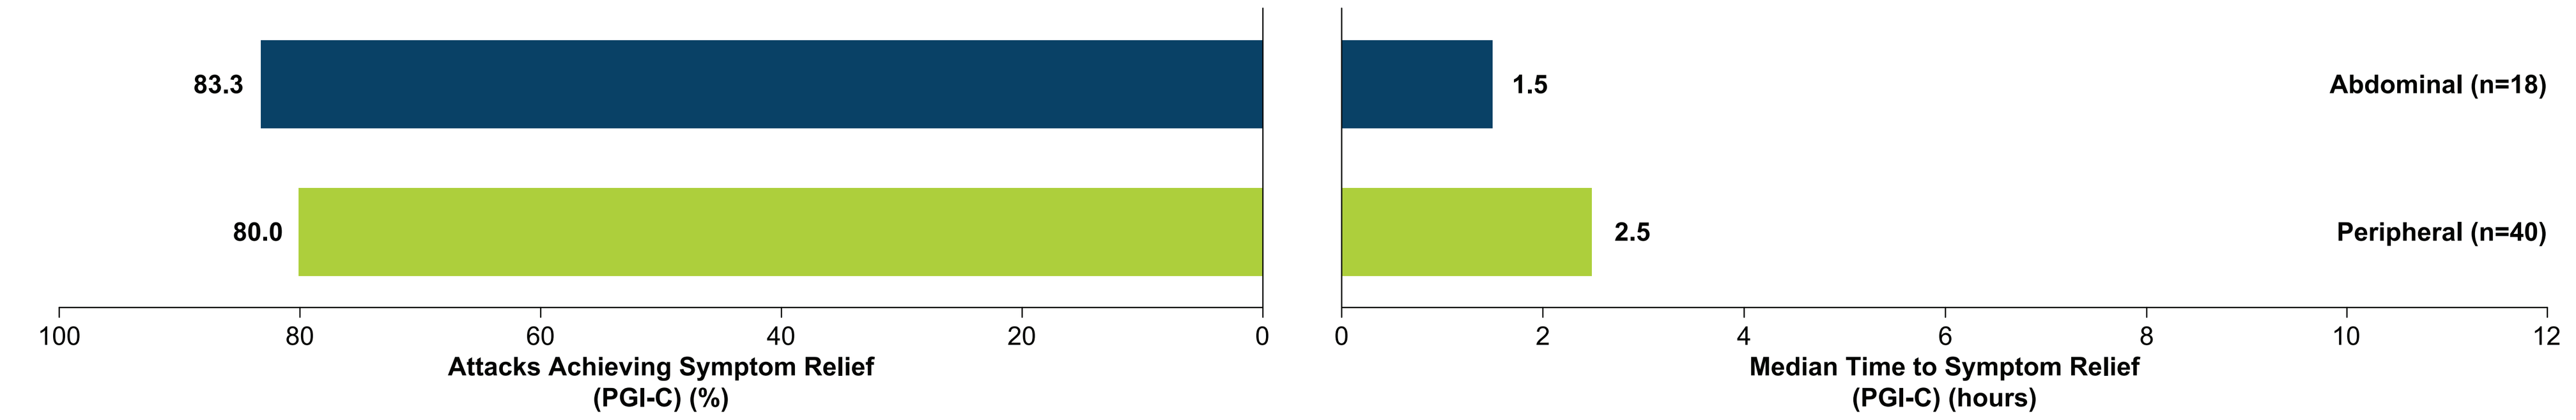

83.3
80.0
1.5
2.5
Abdominal (n=18)
Peripheral (n=40)
100
80
60
40
20
0
Attacks Achieving Symptom Relief (PGI-C) (%)
0
2
4
6
8
10
12
Median Time to Symptom Relief (PGI-C) (hours)

Symptom relief was defined as a score of "A Little Better" or higher for 2 consecutive timepoints within 12 hours of study drug administration on the PGI-C scale.
PGI-C, Patient Global Impression of Change.

- 66.7% of abdominal attacks and 50.0% of peripheral attacks improved on PGI-S within 12 hours (**Figure 5, left panel**)
- Based on PGI-S, median time to improvement within 12 hours was shorter for abdominal attacks (**Figure 5, right panel**)

Figure 5. Achievement of Symptom Improvement and Time to Symptom Improvement (PGI-S) Within 12 Hours by Attack Location

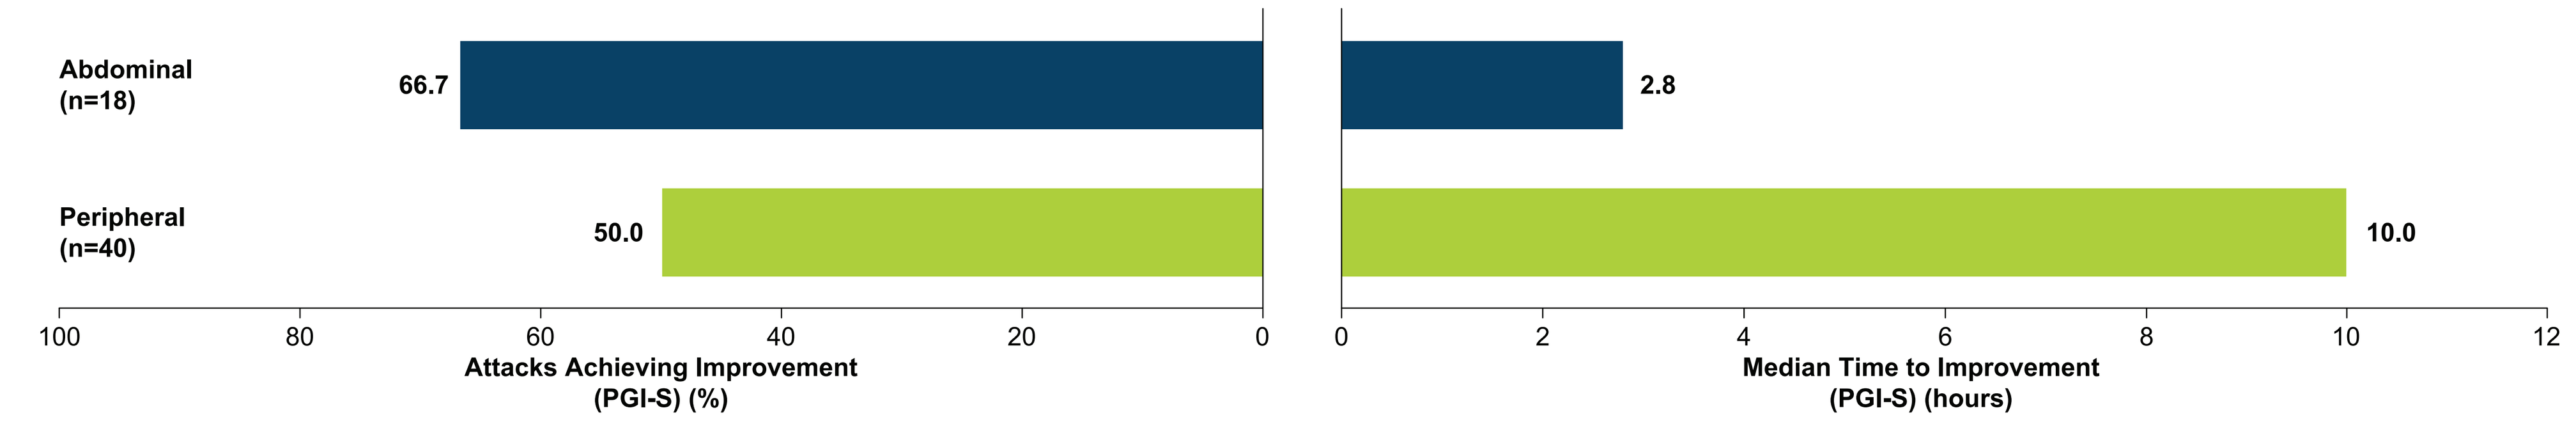

Abdominal (n=18)
Peripheral (n=40)
66.7
50.0
2.8
10.0
100
80
60
40
20
0
Attacks Achieving Improvement (PGI-S) (%)
0
2
4
6
8
10
12
Median Time to Improvement (PGI-S) (hours)

Improvement on PGI-S scale was defined as improvement from baseline by 1 or more levels within 12 hours.
PGI-S, Patient Global Impression of Severity.

Attack Resolution

- More abdominal (61.1%) than peripheral (50.0%) attacks achieved resolution within 24 hours on PGI-S (**Figure 6, left panel**)
- Median time to attack resolution (PGI-S) was 9 and 21 hours for abdominal and peripheral attacks, respectively (**Figure 6, right panel**)

Figure 6. Resolution of Attacks and Time to Attack Resolution (PGI-S) Within 24 Hours by Attack Location

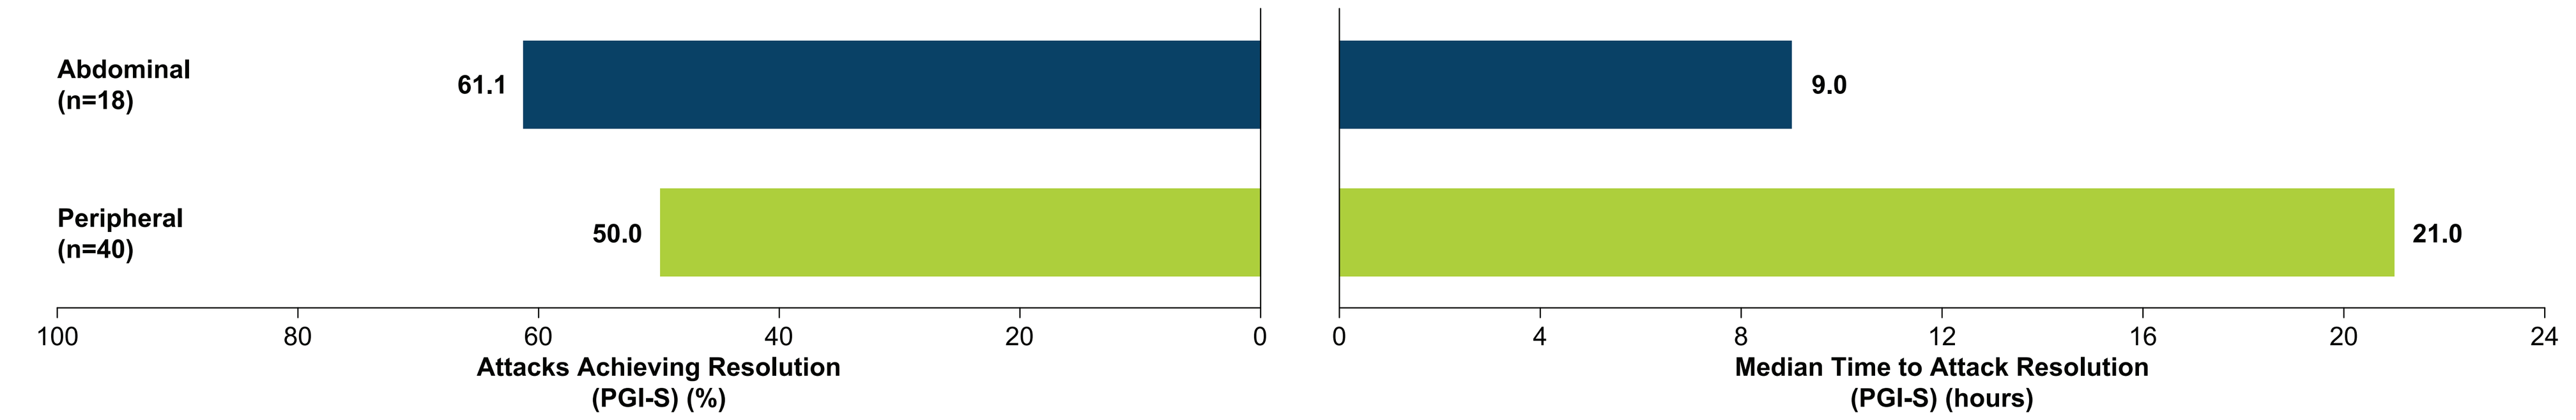

Abdominal (n=18)
Peripheral (n=40)
61.1
50.0
9.0
21.0
100
80
60
40
20
0
Attacks Achieving Resolution (PGI-S) (%)
0
4
8
12
16
20
24
Median Time to Attack Resolution (PGI-S) (hours)

Attack resolution was defined as a PGI-S score of 0 ("None") within 24 hours.
PGI-S, Patient Global Impression of Severity.

- More abdominal (76.5%) than peripheral (60.6%) attacks achieved resolution within 24 hours on VAS (**Figure 7, left panel**)
- Median time to attack resolution (VAS) was 6 and 10 hours for abdominal and peripheral attacks, respectively (**Figure 7, right panel**)

Figure 7. Resolution of Attacks and Time to Attack Resolution (VAS) Within 24 Hours by Attack Location

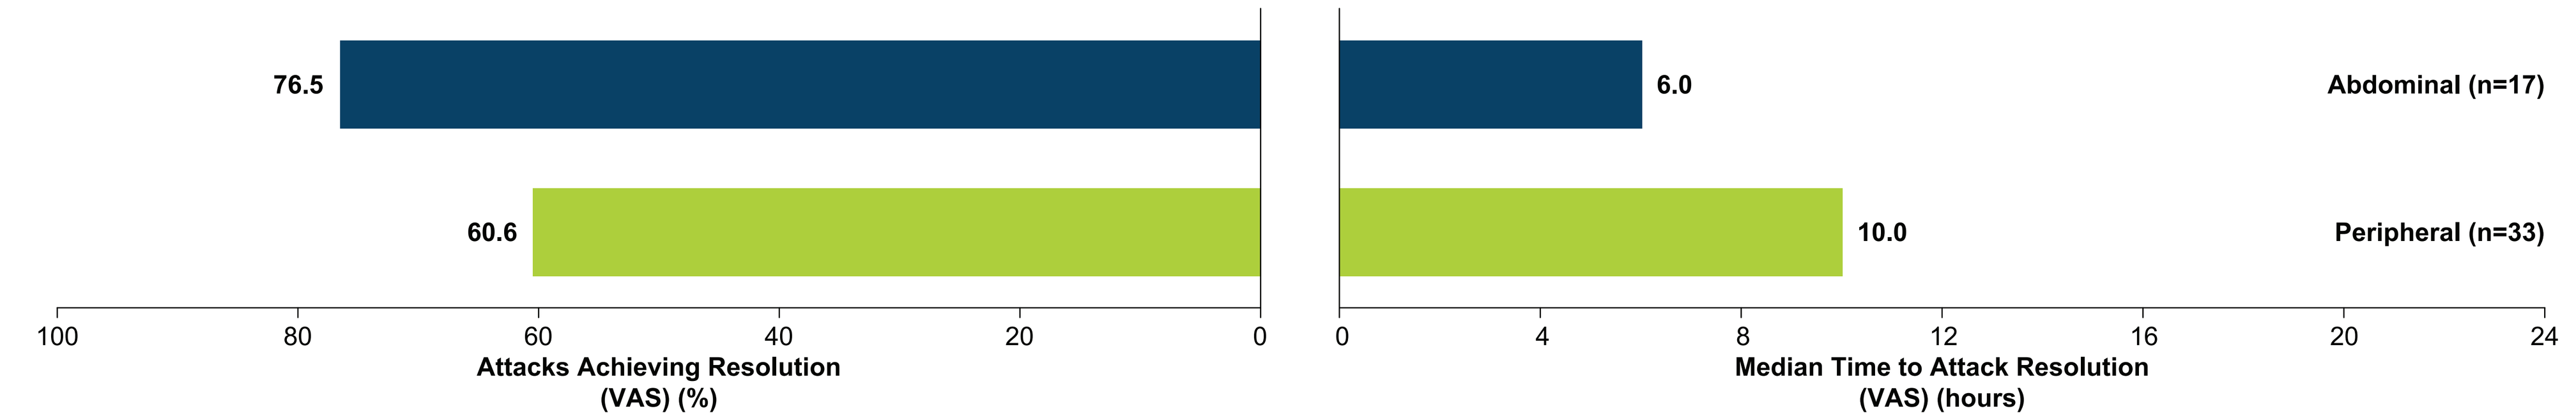

76.5
60.6
6.0
10.0
Abdominal (n=17)
Peripheral (n=33)
100
80
60
40
20
0
Attacks Achieving Resolution (VAS) (%)
0
4
8
12
16
20
24
Median Time to Attack Resolution (VAS) (hours)

Attack resolution was defined as a score <10 mm for all VAS components for 3 consecutive timepoints within 24 hours.
VAS, visual analog scale.

Rescue Medication Use

- For attacks treated with sebetralstat, rates of rescue medication use were low within 12 and 24 hours of study drug administration (**Figure 8, both panels**)

Figure 8. Rescue Medication Use Within 12 and 24 Hours by Attack Location

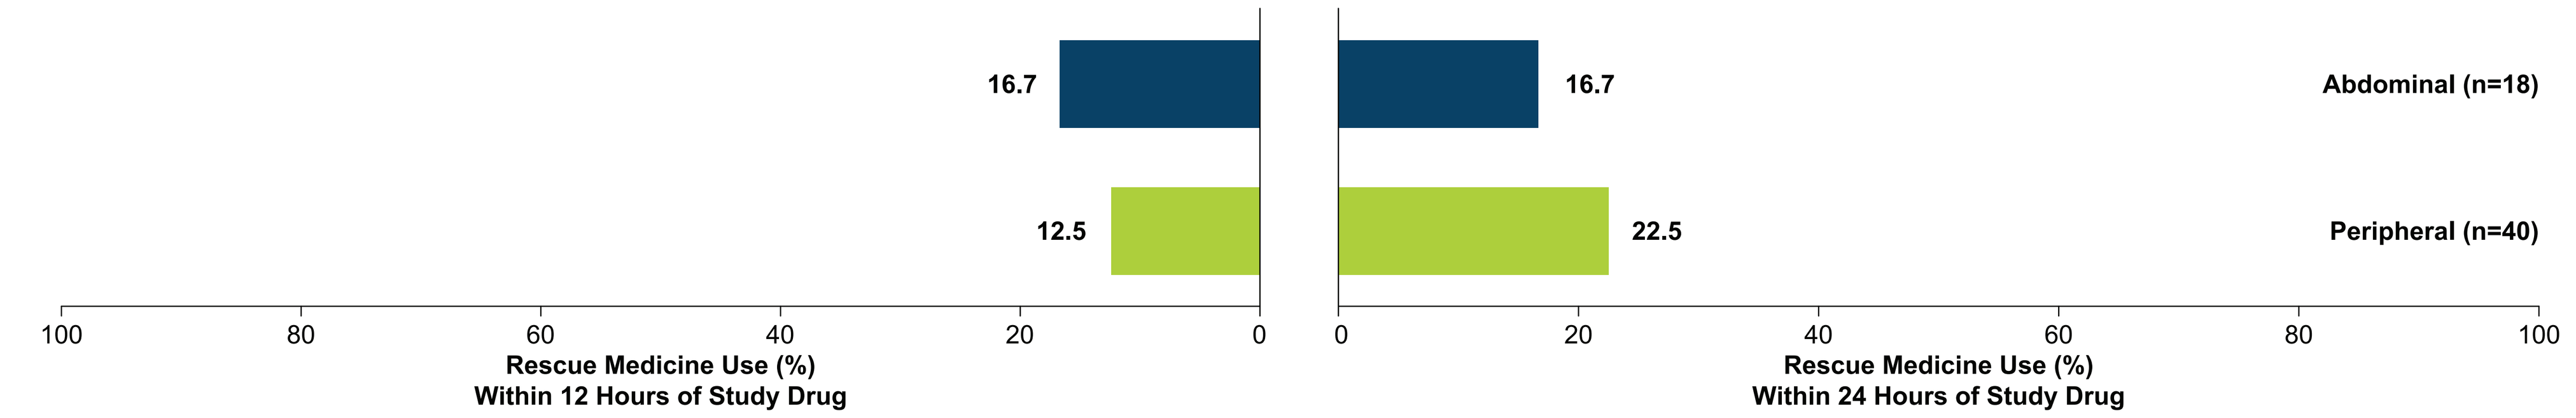

16.7
12.5
16.7
22.5
Abdominal (n=18)
Peripheral (n=40)
100
80
60
40
20
0
Rescue Medicine Use (%) Within 12 Hours of Study Drug
0
20
40
60
80
100
Rescue Medicine Use (%) Within 24 Hours of Study Drug

Conclusions

- Sebetralstat treatment resulted in rapid symptom relief for both abdominal and peripheral attacks, with approximately 80% achieving symptom relief within 12 hours regardless of attack location as evaluated by a score of "A Little Better" or higher for 2 consecutive timepoints on the PGI-C scale
 - Abdominal attacks tended to resolve more quickly than peripheral attacks as evaluated on PGI-S and VAS scales
 - Faster symptom relief for abdominal versus peripheral attacks observed in this study is consistent with previously reported findings for other acute therapies[13,14]
 - The use of rescue medication for abdominal and peripheral attacks treated by sebetralstat was also similar, although somewhat more abdominal attacks were rescued earlier, while more peripheral attacks were rescued later
- The results of this post hoc analysis demonstrate that sebetralstat provides symptom relief and attack resolution for people living with HAE, regardless of abdominal or peripheral attack location

Acknowledgments

Medical writing assistance was provided under the direction of the authors by Katherine Stevens-Favorite, PhD, of Cadent, a Syneos Health group company, and was supported by KalVista Pharmaceuticals, Inc.

Presented during the Western Society of Allergy, Asthma and Immunology (WSAAI) 60th Annual Scientific Session, February 5-9, 2023, Kamuela, HI, US.

Disclosures

EAP was the principal investigator and reviewed and approved the study report. AZ has received speaker/consultancy fees from BioCryst, CSL Behring, Pharming, and Takeda. DMC has received speaker fees and/or consultancy fees from BioCryst, CSL Behring, Ionis Pharmaceuticals, KalVista, Pharming, Pharvaris, and Shire/Takeda. PW is an employee of Veramed Limited, and acts as a consultant statistician for KalVista. PKA, CMY, and MDS are employees of KalVista Pharmaceuticals.

References

1. Bork K, et al. *Am J Med*. 2006;119(3):267-274.
2. Longhurst H, Cicardi M. *Lancet*. 2012;379(9814):474-481.
3. Banerji A, et al. *N Engl J Med*. 2017;376(8):717-728.
4. Schmaier AH. *Front Med*. 2018;5:3.
5. Busse PJ, et al. *J Allergy Clin Immunol Pract*. 2021;9(1):132-150.e3.
6. Maurer M, et al. *Allergy*. 2018;73(8):1575-1596.
7. Maurer M, et al. *Allergy*. Published online January 10, 2022. doi:10.1111/all.15214
8. Kalbitor. Package insert. Takeda Pharmaceuticals America, Inc; 2019.
9. Berinert. Package insert. CSL Behring; 2019.
10. Ruconest. Package insert. Pharming; 2014.
11. Firazyr. Package insert. Takeda Pharmaceuticals America, Inc.; 2020.
12. Aygören-Pürsün E, et al. *Lancet*. In press.
13. Craig TJ, et al. *Allergy*. 2011;66(12):1604-1611.
14. Riedl M, et al. *Ann Allergy Asthma Immunol*. 2010;105(6):430-436.e2.